A Multi-Center Validation Study of Quantitative Imaging Mass Spectrometry

Jeremy A. Barry;[1] Rima Ait-Belkacem;[2] William M. Hardesty;[1] Lydia Benakli;[2] Juliette Masure; [2] David Bonnel; [2] Jonathan Stauber;[2,3] Stephen Castellino[1]

[1] *Bioimaging, GlaxoSmithKline, Collegeville, PA, United States*

[2] *Research and Development, Imabiotech Corp, Loos, France*

[3] *Research and Development, Imabiotech Corp, Billerica, MA, United States*

Introduction

Over the last decade, considerable effort has been directed towards the development and evolution of quantitative imaging mass spectrometry (IMS).[1] These capabilities have most notably been applied to the quantitative evaluation of drug distribution in sections of tissue following dosing. Knowledge of the distribution alone can be largely beneficial but understanding the distribution on a quantified scale can have significant implications for drug efficacy or, in some cases, toxicity.[2]

Several key approaches the quantitative IMS experiment have emerged which are nicely summarized by Bonnel and Porta.[3-4] The approaches to constructing the calibration curve for quantitative IMS can be categorized as the in-solution, on-tissue, and in-tissue methods. Each comes with distinct advantages and drawbacks. The calibration curve is spotted on the sample target adjacent to the tissue to be quantified in the in-solution method. While straightforward in practice, without an internal standard this approach often fails to account for matrix effects such as extraction efficiency and ion suppression. The on-tissue methods are similar in practice to the in-solution approach but the calibration curve is spotted on a control tissue section. However, heterogeneity within the control tissue section can skew the calibration. The in-tissue approach, also known as the mimetic tissue model, utilizes a standard curve which is spiked in tissue homogenates.[5] While this method more closely replicates the conditions of the analyte in the sample tissue, it was often remarked that the preparation was too involved. A recently published protocol outlines a streamlined approach to the mimetic tissue model which addresses these comments.[6]

Despite consistent efforts to substantiate the legitimacy of the quantitative capabilities of IMS through validation with more established methods like LC-MS, questions still remain as to the level of variation that can be expected from the IMS experiment. Through a multi-center study, we aim to address these concerns by assessing the accuracy and precision of quantitative IMS using the two most commonly used methodologies, the on-tissue dilution series and the mimetic tissue model. In addition, the validity of normalization to a homogenously coated stable isotopically-labeled (SIL) internal standard is also evaluated for both approaches. In this study three analysts from different sites acquired quantitative IMS data from a common sample using a standardized protocol.

Study Outline

A schematic outlining the multi-center study is provided in **Figure 1**. The right median lobe of a rat liver was perfused with clozapine (CLZ) was frozen and portioned into two tissue blocks and distributed for IMS analysis by Imabiotech and GlaxoSmithKline. A protocol was distributed with the samples outlining the key aspects of sample preparation, acquisition, and data analysis. In short, each analyst prepared a mimetic tissue model and dilution series solutions for quantification of CLZ and its primary metabolite norclozapine (NCLZ).

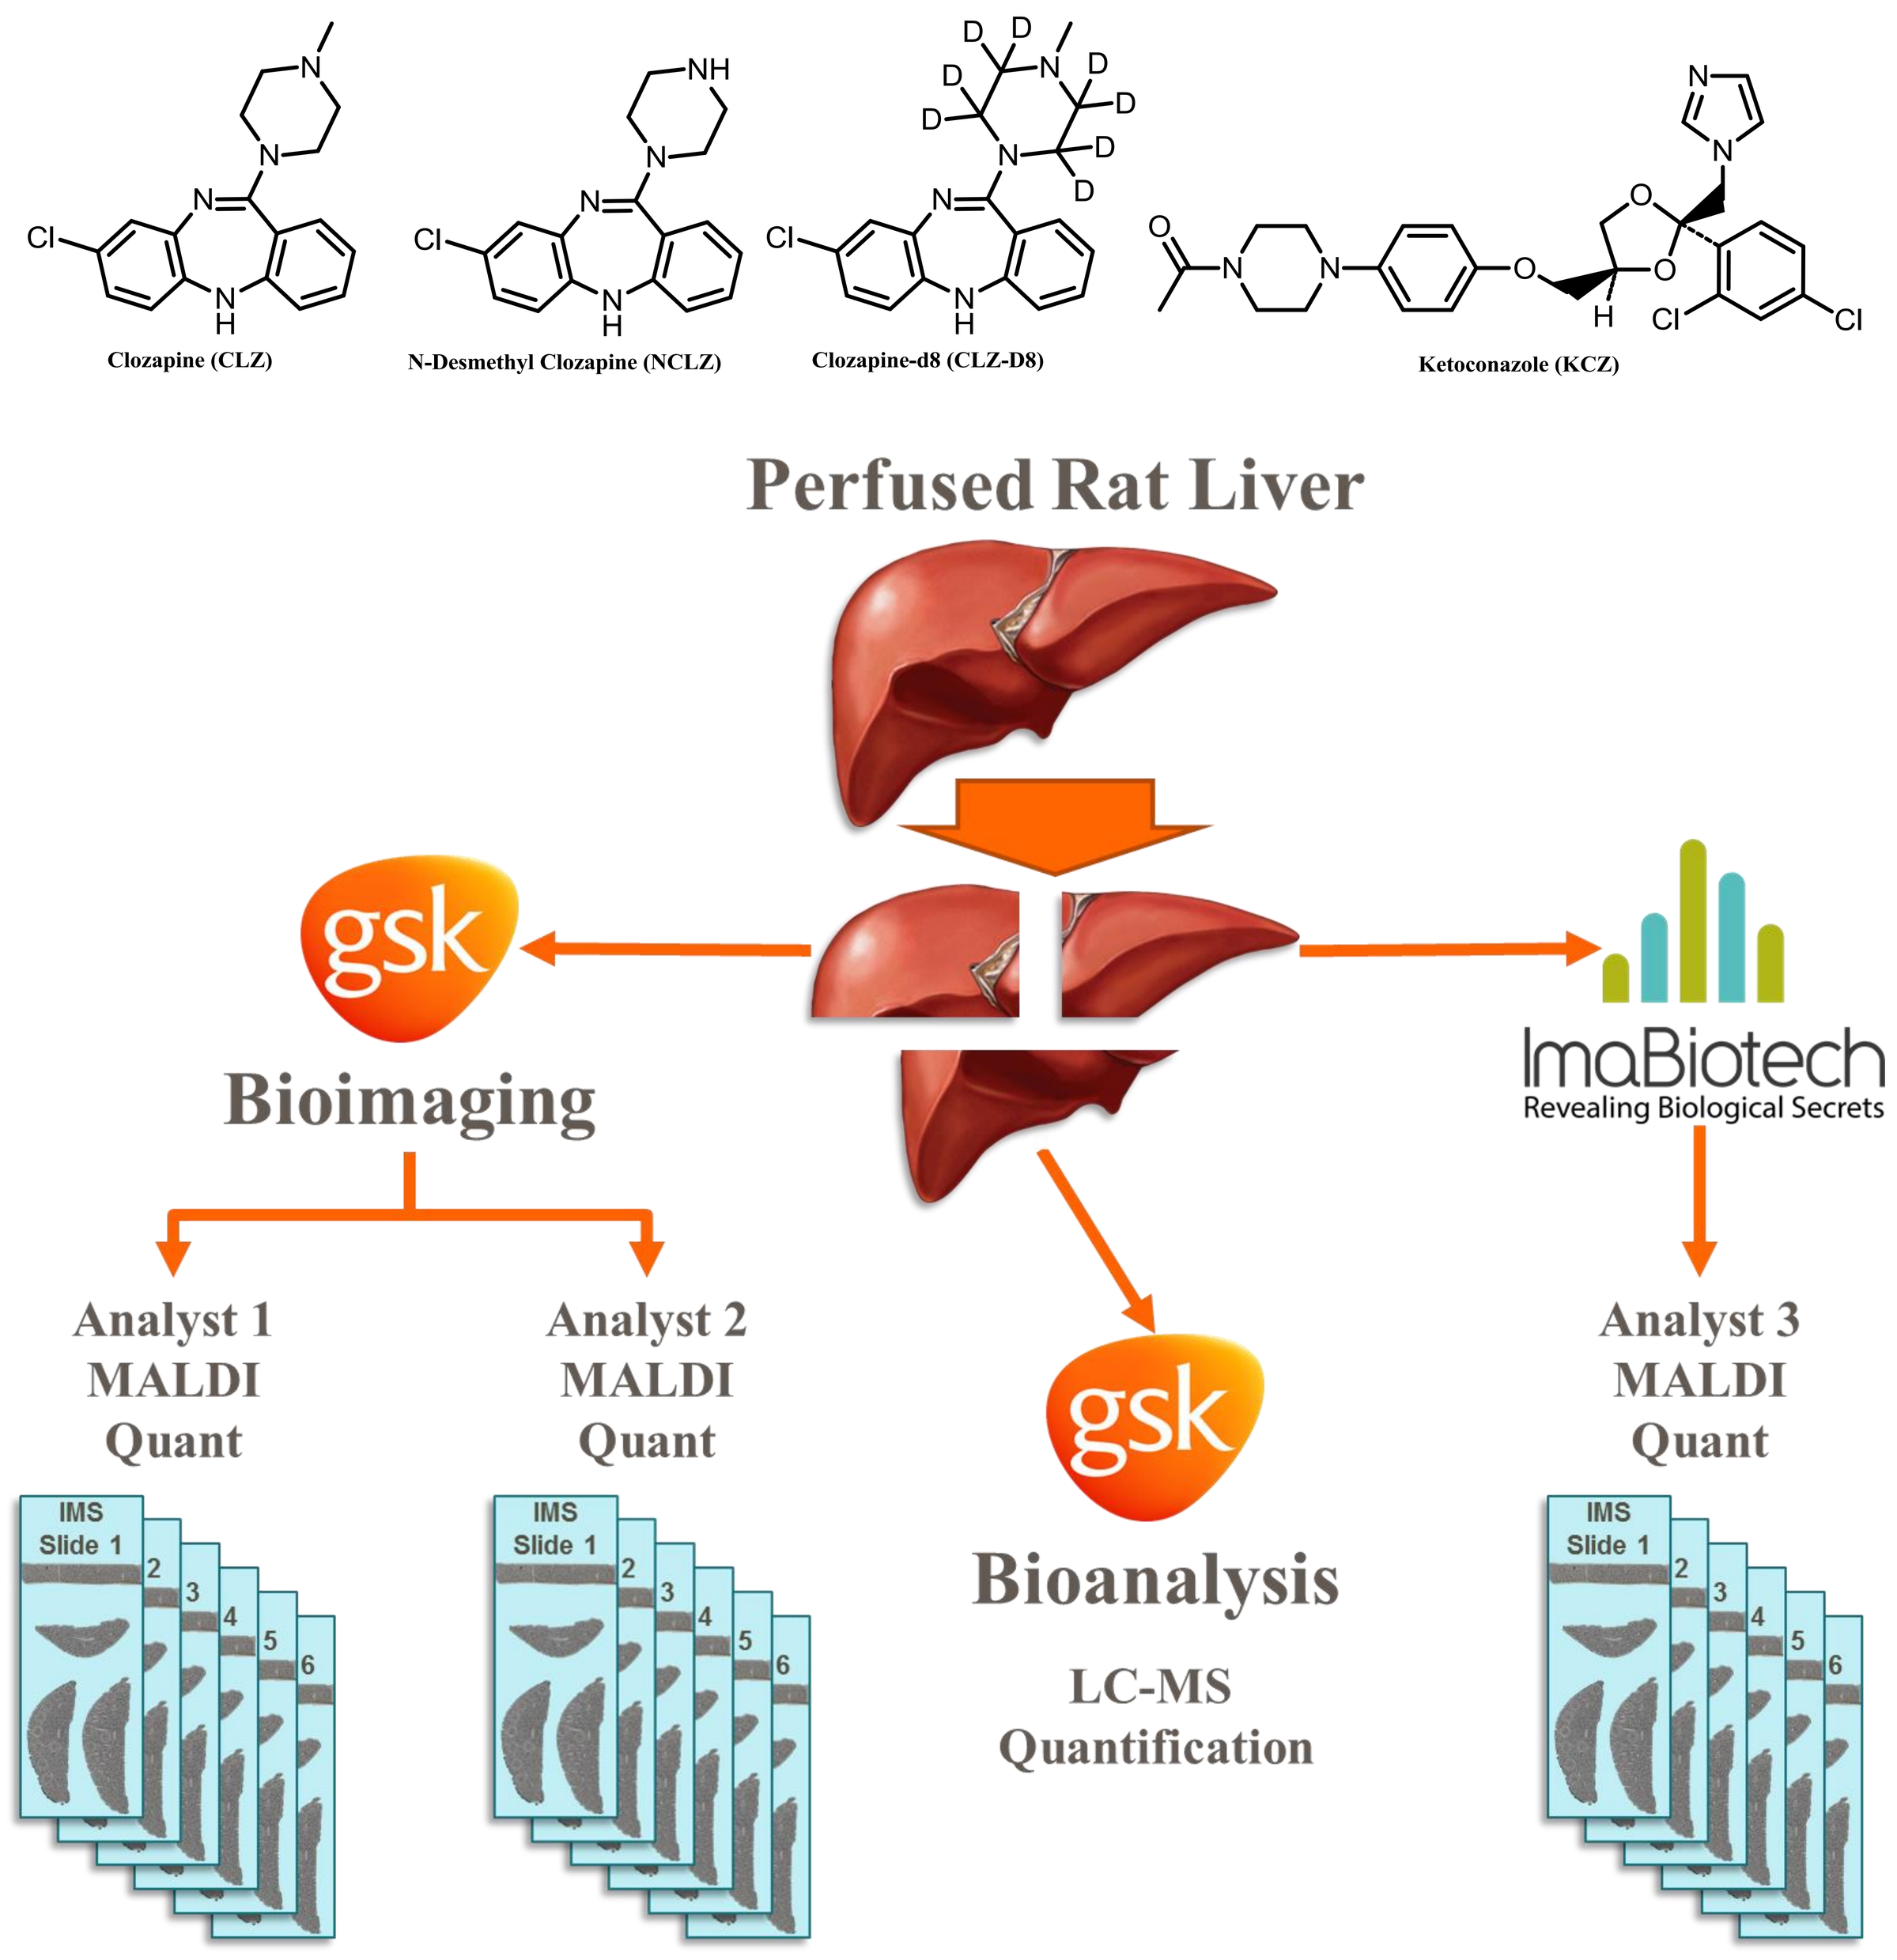
Clozapine (CLZ)
N-Desmethyl Clozapine (NCLZ)
Clozapine-d8 (CLZ-D8)
Ketoconazole (KCZ)
Perfused Rat Liver
Bioimaging
ImaBiotech
Revealing Biological Secrets
Analyst 1 MALDI Quant
Analyst 2 MALDI Quant
Analyst 3 MALDI Quant
Bioanalysis
LC-MS Quantification
IMS Slide 1

Figure 1: Outline of the Multi-Site Validation Study
Tissue blocks from a tissue perfused with clozapine (CLZ) were distributed for analysis by quantitative IMS on three sites and sections from the block were quantified by LC-MS to act as a target value for assessment of quantitative accuracy.

An example acquisition is provided for reference in **Figure 2** to show the layout of the mimetic tissue model, quality control samples, IPRL tissue, and the dilution series. The mimetic tissue model contained seven levels. Six of the levels were spiked with CLZ and NCLZ to give an inverse gradient of the two analytes. The last layer represented a blank tissue measurement for both CLZ and NCLZ calibration curves but was spiked with ketoconazole (KCZ) which was used as a pre-run and post-run quality control sample. The analysts prepared six replicate slides for IMS analysis each containing a section of the IPRL tissue, a section of the mimetic tissue model, and control liver tissue sections for the dilution series. The tissue sections were thaw-mounted onto ITO coated microscope slides and stored at -80 ° C. Each of the six slides were then individually prepared on the day of analysis just prior to IMS acquisition. The dilution series was spotted onto the control tissue sections and the slides were coated with DHB spiked with CLZ-d8 using a pre-defined method on an HTX TM Sprayer. At each of the three sites, IMS was acquired on a Bruker solariX FT-ICR mass spectrometer. Several key parameters of the IMS acquisition method were kept constant across each analysis where other acquisition method parameters were allowed to be optimized per individual instrument. Serial sections of the IPRL tissue were collected, homogenized, and quantified using LC-MS by an independent group at GSK. This quantification was used as the target value for determining the accuracy of the IMS quantification.

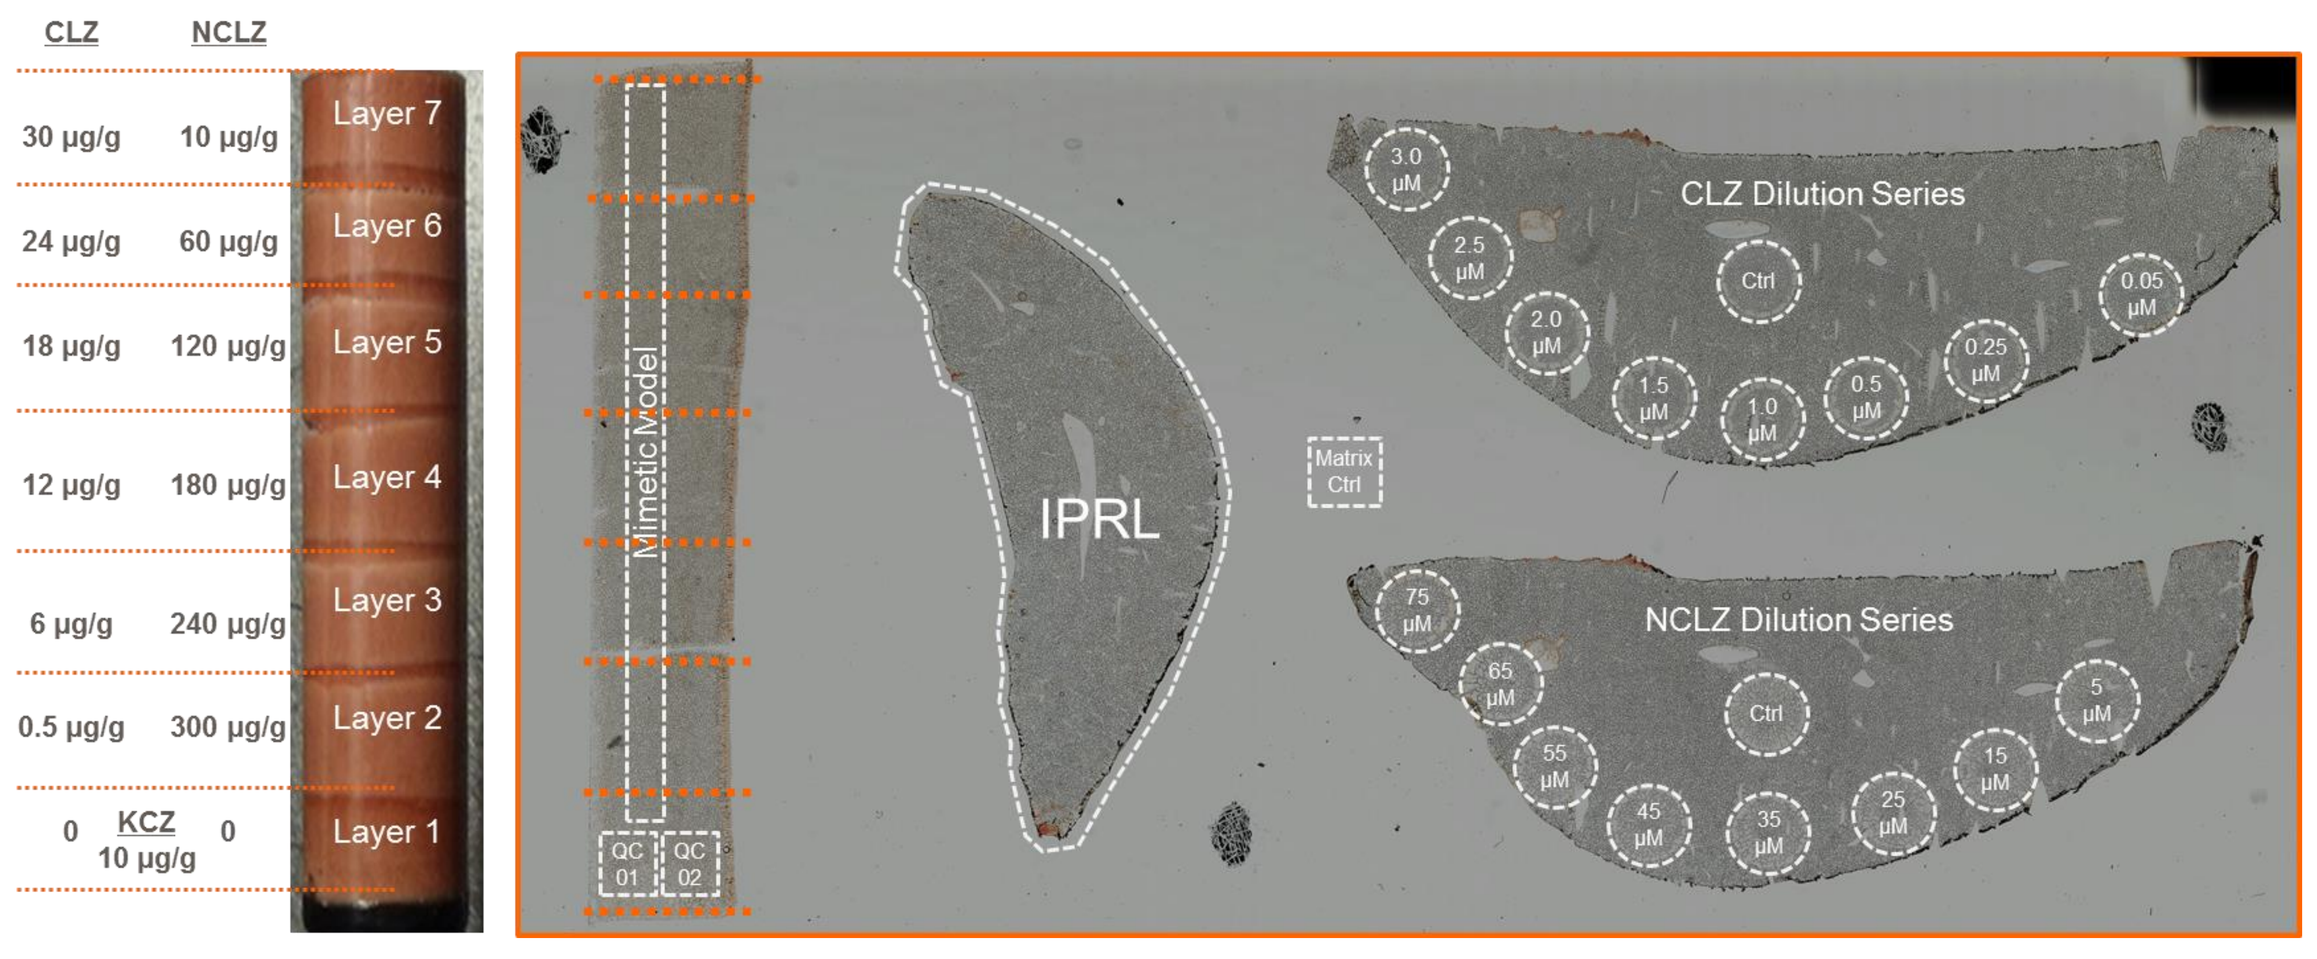
CLZ NCLZ
30 µg/g 10 µg/g Layer 7
24 µg/g 60 µg/g Layer 6
18 µg/g 120 µg/g Layer 5
12 µg/g 180 µg/g Layer 4
6 µg/g 240 µg/g Layer 3
0.5 µg/g 300 µg/g Layer 2
0 KCZ 10 µg/g 0 Layer 1
Mimetic Model
QC 01
QC 02
IPRL
Matrix Ctrl
CLZ Dilution Series
NCLZ Dilution Series

Figure 2: Schematic of the IMS Acquisition
Provides a description of the various levels of the mimetic tissue model with quality control regions. Additionally shows an example of the IMS acquisition of each of the pertinent areas for the mimetic tissue model and dilution series quantification methods.

Quality Control

Quality control samples were run before and after each acquisition as a way to evaluate within run and between run variability. The quality control sample was a tissue homogenate spiked with ketoconazole at a level of 10 µg/g. There were no clear trends across the three analysts in regards to improvement or reduction of signal neither between nor within runs but the RSD of the signal intensity was consistent at around 35%.

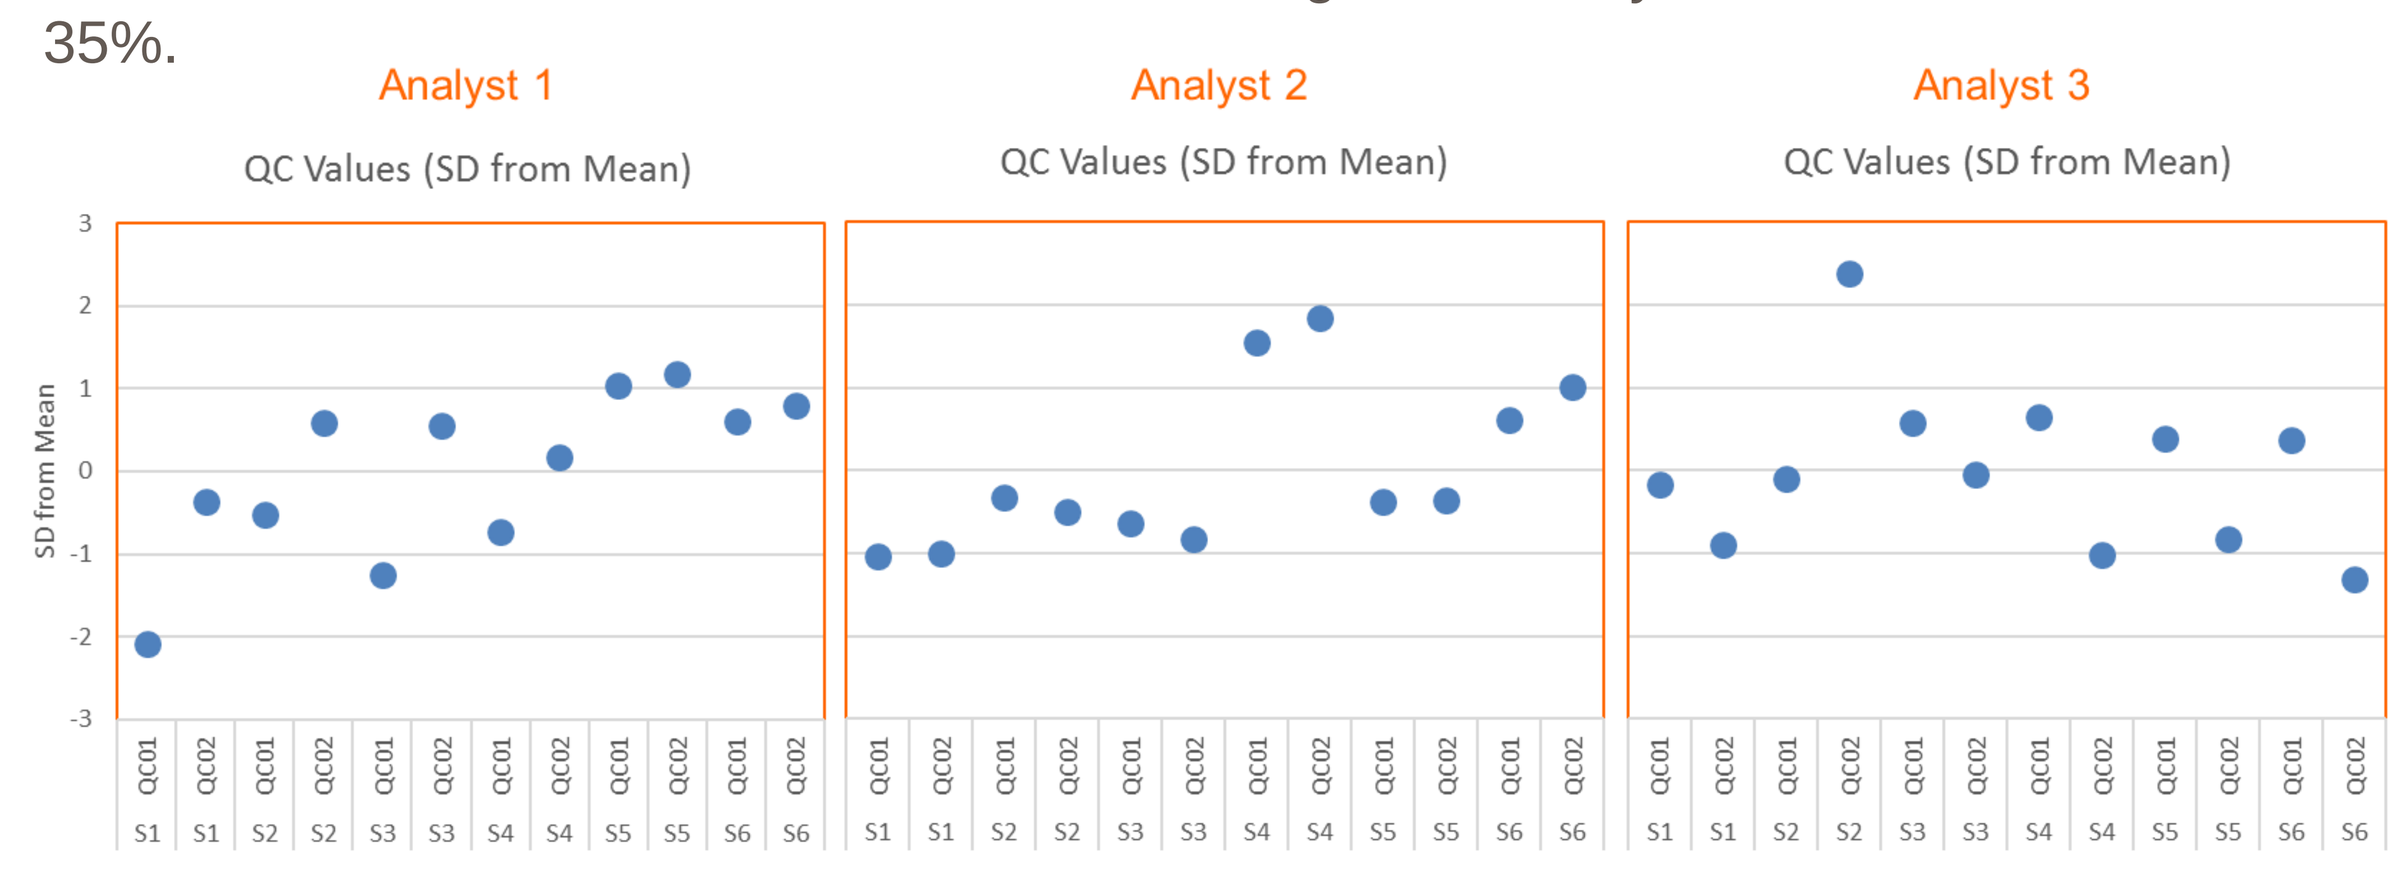
Analyst 1
Analyst 2
Analyst 3
QC Values (SD from Mean)
SD from Mean

	Analyst 1 All	Analyst 1 Pre-run	Analyst 1 Post-run	Analyst 2 All	Analyst 2 Pre-run	Analyst 2 Post-run	Analyst 3 All	Analyst 3 Pre-run	Analyst 3 Post-run
RSD	35%	16%	49%	34%	32%	38%	35%	10%	51%

Figure 3: IMS Quantification Quality Control
A quality control sample was evaluated before and after each acquisition in an attempt to characterize within and between run variability.

Repeatability

A key metric for quantification is the ability to consistently obtain the same or similar results upon repetition of the method. Applied to imaging mass spectrometry, it is somewhat difficult to truly assess as samples typically cannot be re-analyzed. To test this capability would require collecting serial sections. While it is expected that the results from serial sections should be largely equivalent, each section is inherently biologically unique. **Figure 4** shows the IMS quantification results for the mimetic tissue model and dilution series both with and without normalization grouped by analyst. Also shown is the LC-MS target value. These results are also provided in **Table 1**. From these results the mimetic tissue model yielded a tighter distribution of predicted concentrations for the six replicates analyzed by each analyst.

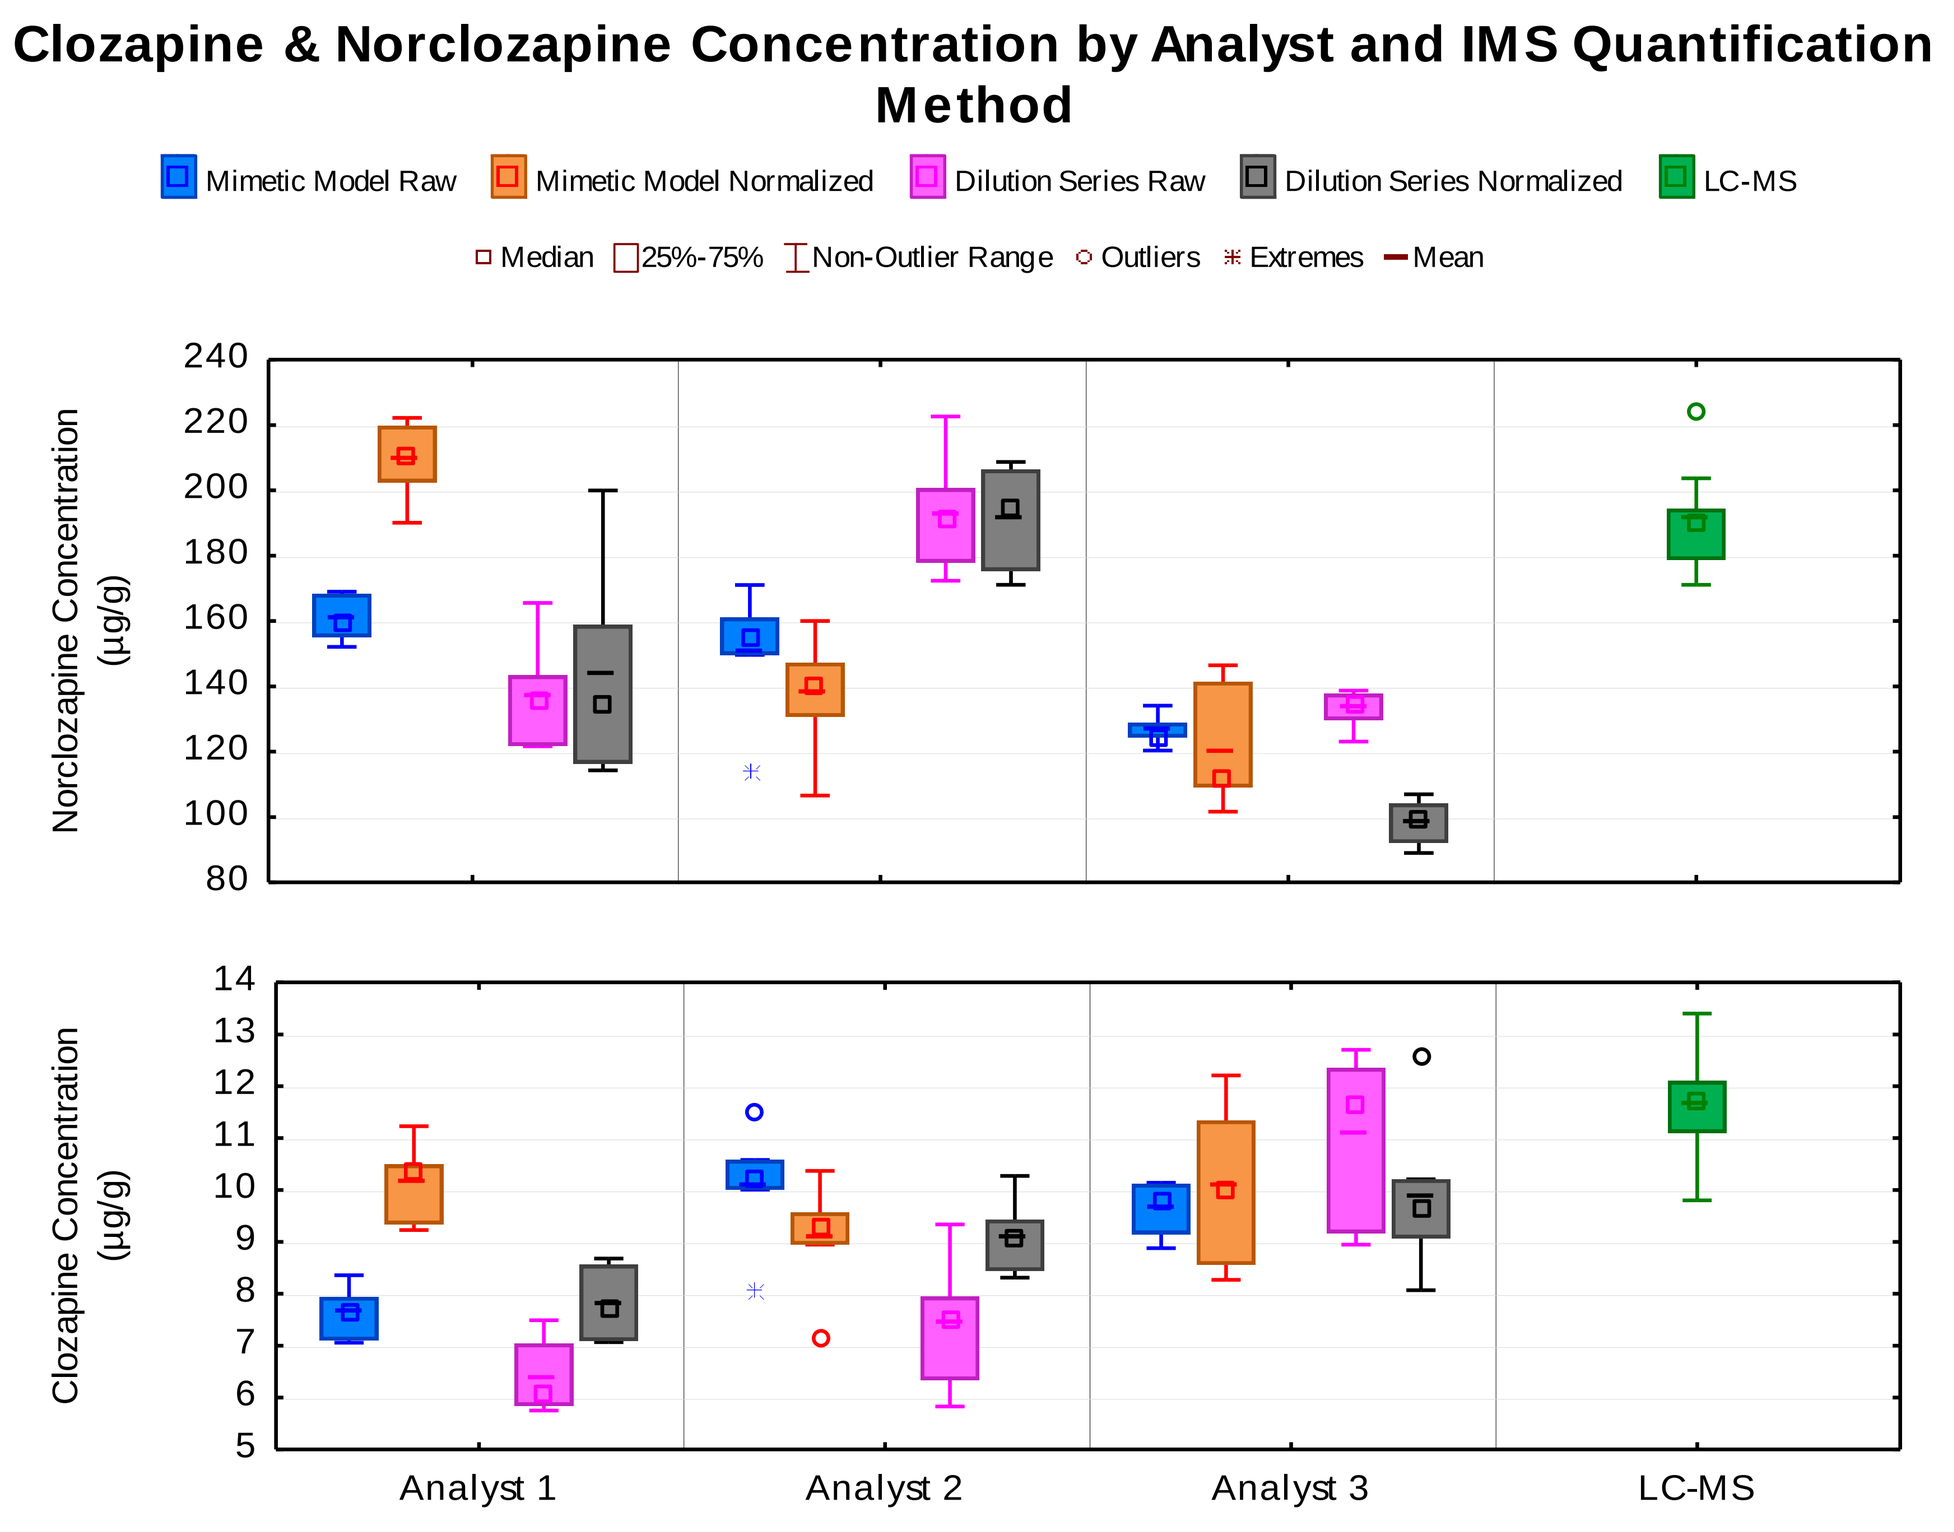
Clozapine & Norclozapine Concentration by Analyst and IMS Quantification Method
Mimetic Model Raw
Mimetic Model Normalized
Dilution Series Raw
Dilution Series Normalized
LC-MS
Median
25%-75%
Non-Outlier Range
Outliers
Extremes
Mean
Norclozapine Concentration (µg/g)
Clozapine Concentration (µg/g)
Analyst 1
Analyst 2
Analyst 3
LC-MS

Figure 4: IMS Quantification Grouped by Analyst
Box plots of the six replicates for each analyst are provided and provide an indication of the precision and accuracy when compared to the LC-MS results. In general, the precision and accuracy of the mimetic model were consistently better compared to the dilution series. The impact of the normalization to a homogenously coated internal standard was inconsistent between analysts and across the two compounds tested.

		CLZ				NCLZ			
		Mimetic Model		Dilution Series		Mimetic Model		Dilution Series	
		Raw	Norm	Raw	Norm	Raw	Norm	Raw	Norm
Analyst 1	S1	7.94	10.18	5.84	8.68	168.99	215.39	141.60	199.99
	S2	8.36	11.23	7.49	8.56	168.29	219.76	143.32	138.28
	S3	7.63	10.48	6.07	8.13	156.86	206.03	130.00	158.77
	S4	7.62	9.23	6.14	7.07	161.04	190.12	121.68	130.76
	S5	7.06	10.49	5.75	7.09	152.11	222.19	121.80	116.41
	S6	7.1	9.34	7.04	7.35	155.09	202.46	165.55	114.28
Analyst 2	S1	10.01	9.57	5.83	9.16	161.04	160.05	177.89	208.70
	S2	10.08	8.95	7.78	10.27	157.71	147.27	189.61	191.15
	S3	11.47	10.37	6.34	8.44	171.05	136.71	193.20	206.38
	S4	10.31	9	9.34	8.31	151.5	130.69	222.62	197.29
	S5	8.08	7.14	7.26	9.43	114.52	106.58	172.34	175.32
	S6	10.58	9.52	7.95	8.99	149.67	143.7	200.63	171.12
Analyst 3	S1	10.14	12.21	9.17	8.07	120.35	112.93	131.8	106.97
	S2	9.75	9.05	8.95	12.56	125.06	109.08	138.72	103.14
	S3	9.15	11.34	11.93	10.21	124.36	101.66	137.69	95.82
	S4	10.12	10.95	12.7	10.09	124.4	110.06	137.75	104.16
	S5	8.88	8.56	12.35	9.07	128.89	146.48	123.1	92.08
	S6	9.8	8.27	11.39	9.26	134.11	141.4	129.7	89.01
AVG		**9.12**	**9.77**	**8.30**	**8.93**	**145.84**	**155.70**	**154.39**	**144.42**
RSD		**14%**	**13%**	**29%**	**15%**	**13%**	**27%**	**20%**	**30%**
Accuracy		**78%**	**84%**	**71%**	**76%**	**76%**	**82%**	**81%**	**76%**
LC-MS		11.7				190.7			

Table 1: Summary of the IMS Quantification Results

Reproducibility

By combining the data from all three analysts, an assessment of the reproducibility of the mimetic tissue model and the dilution series can be made as well as the impact of normalization to an internal standard. These data are summarized in **Figure 5** and **Table 1**. For CLZ the raw data from the mimetic model was capable of producing precise and relatively accurate data. Normalization slightly improved the precision and accuracy. For the raw dilution series data, the precision and accuracy were relatively poor but normalization improved the data to a level that was comparable to the raw mimetic tissue model data. Each of the IMS methods slightly underestimated the target result provided by LC-MS which could be due to heterogeneity among the samples.

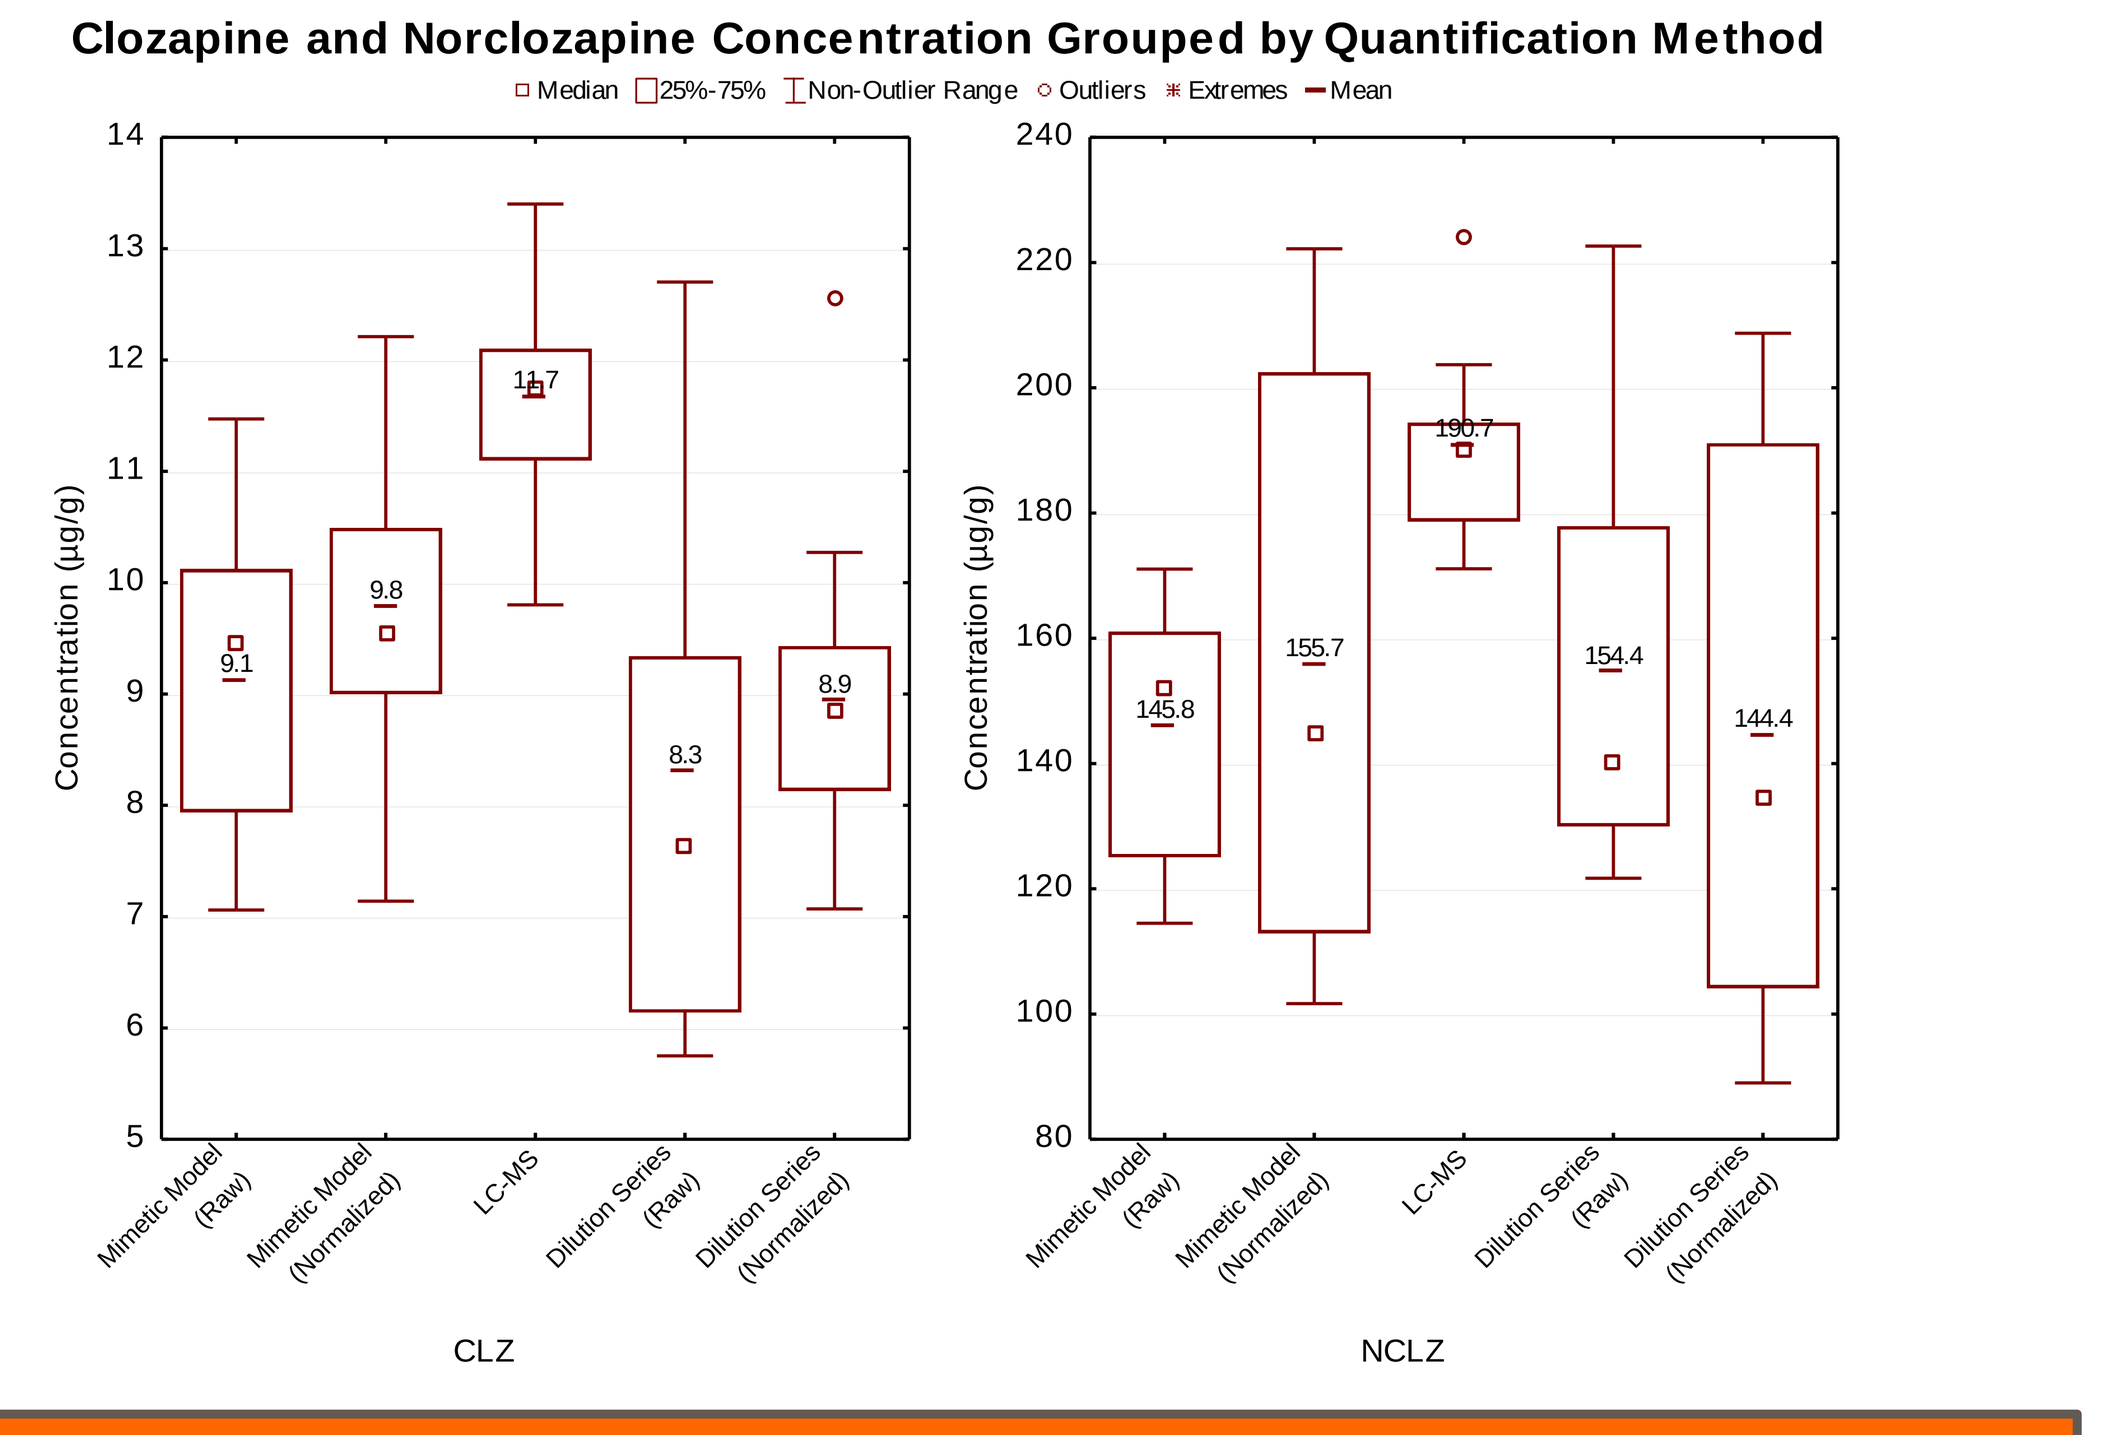
Clozapine and Norclozapine Concentration Grouped by Quantification Method
Median
25%-75%
Non-Outlier Range
Outliers
Extremes
Mean
Concentration (µg/g)
Mimetic Model (Raw)
Mimetic Model (Normalized)
LC-MS
Dilution Series (Raw)
Dilution Series (Normalized)
CLZ
NCLZ

Figure 5: IMS Quantification Grouped by Method
Box plots of the combine the six replicates of each analyst to assess the reproducibility of the IMS quantification. In general, normalization was needed to improve the precision and accuracy of the dilution series to a level which was consistent with the raw mimetic tissue model quantification.

Impact of Normalization to an SIL

Normalization to a homogeneously-coated internal standard is often believed to be necessary for accurate quantification regardless of the method used. **Figure 6** below demonstrated the impact that normalization had on the pixel intensity RSD for each calibration region (**6a**) as well as the accuracy to self-predict the calibrant levels (**6b**). Additionally, the impact on the correlation between ion intensity and concentration for each of the quantification methods is provided (**6c**).

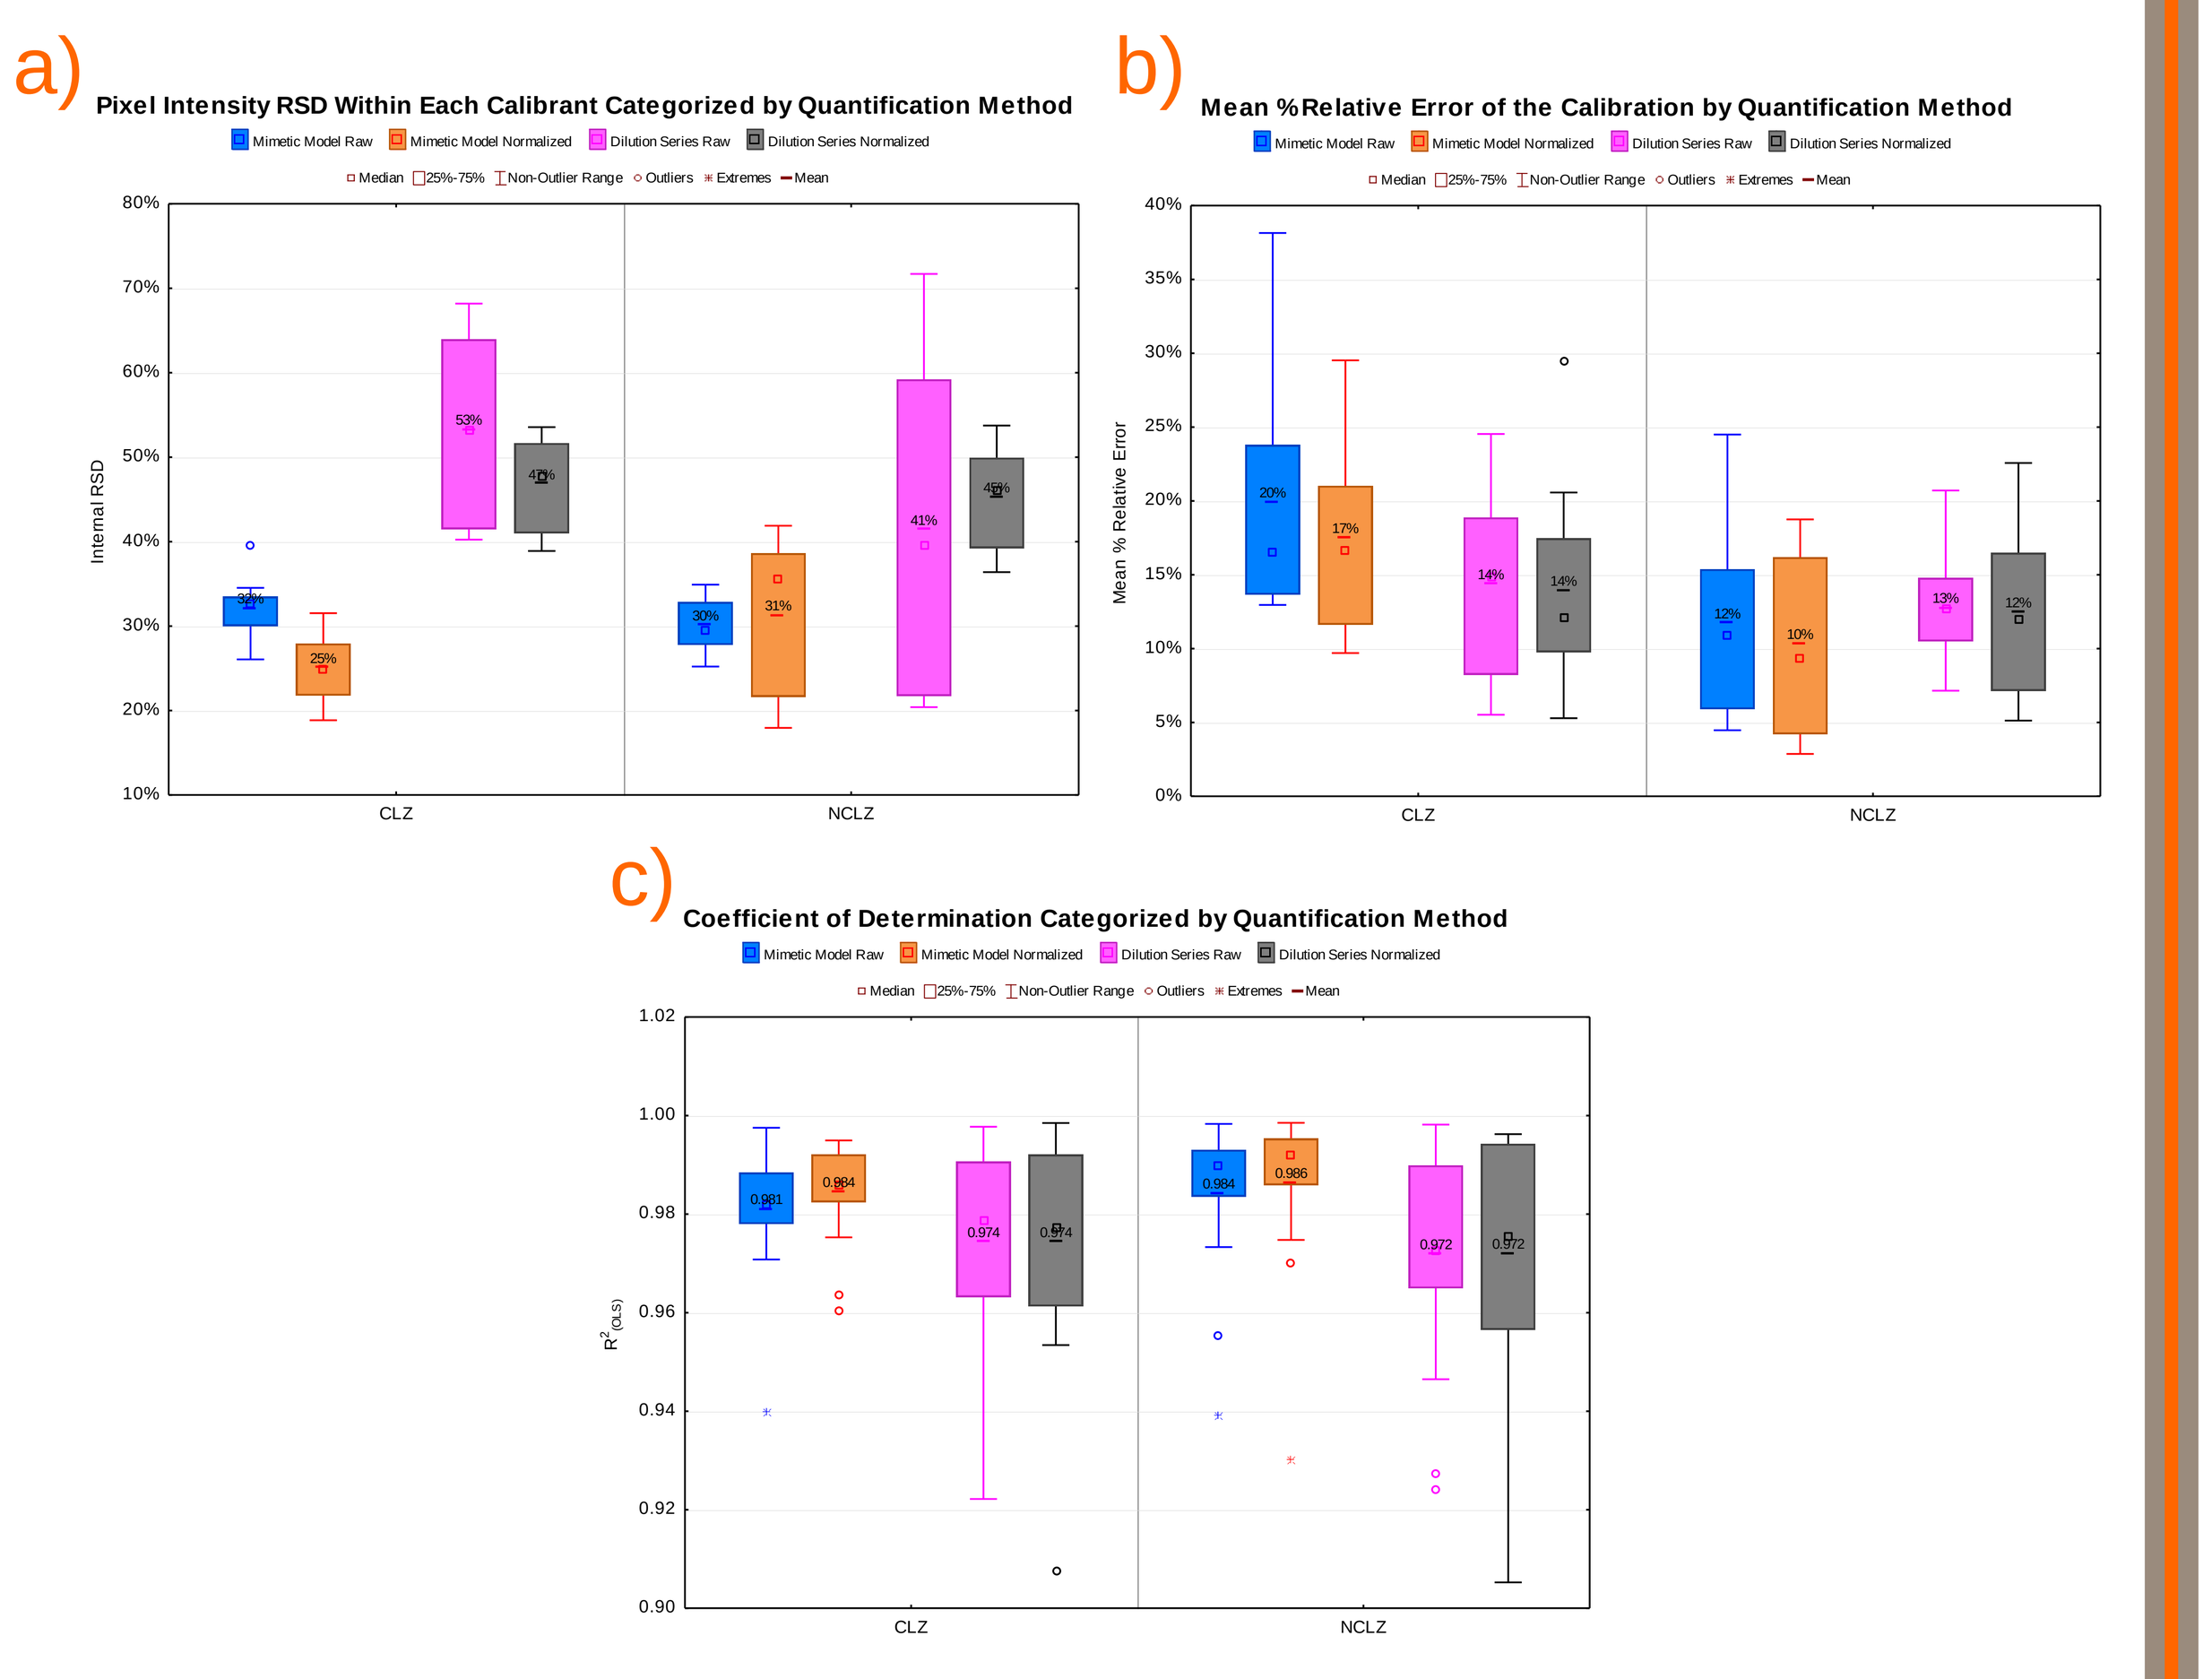
a)
Pixel Intensity RSD Within Each Calibrant Categorized by Quantification Method
Internal RSD
b)
Mean %Relative Error of the Calibration by Quantification Method
Mean % Relative Error
c)
Coefficient of Determination Categorized by Quantification Method
Mimetic Model Raw
Mimetic Model Normalized
Dilution Series Raw
Dilution Series Normalized
CLZ
NCLZ

Figure 6: Impact of Normalization on Internal Precision, Accuracy, and Correlation
6a shows there was inherently less variability within the mimetic tissue model calibrant regions and that normalization generally improved the precision. 6b shows the mean relative error across the calibrant regions for each quantification method. This provides an estimate of how well the resulting calibration can predict the levels of each calibrant. 6c shows the impact of normalization on the coefficient of determination (R^2) for the standard curves of each calibration method. In general, the spread of the correlation coefficients was smaller for the mimetic tissue model and normalization tended to improve the correlation between ion intensity and concentration.

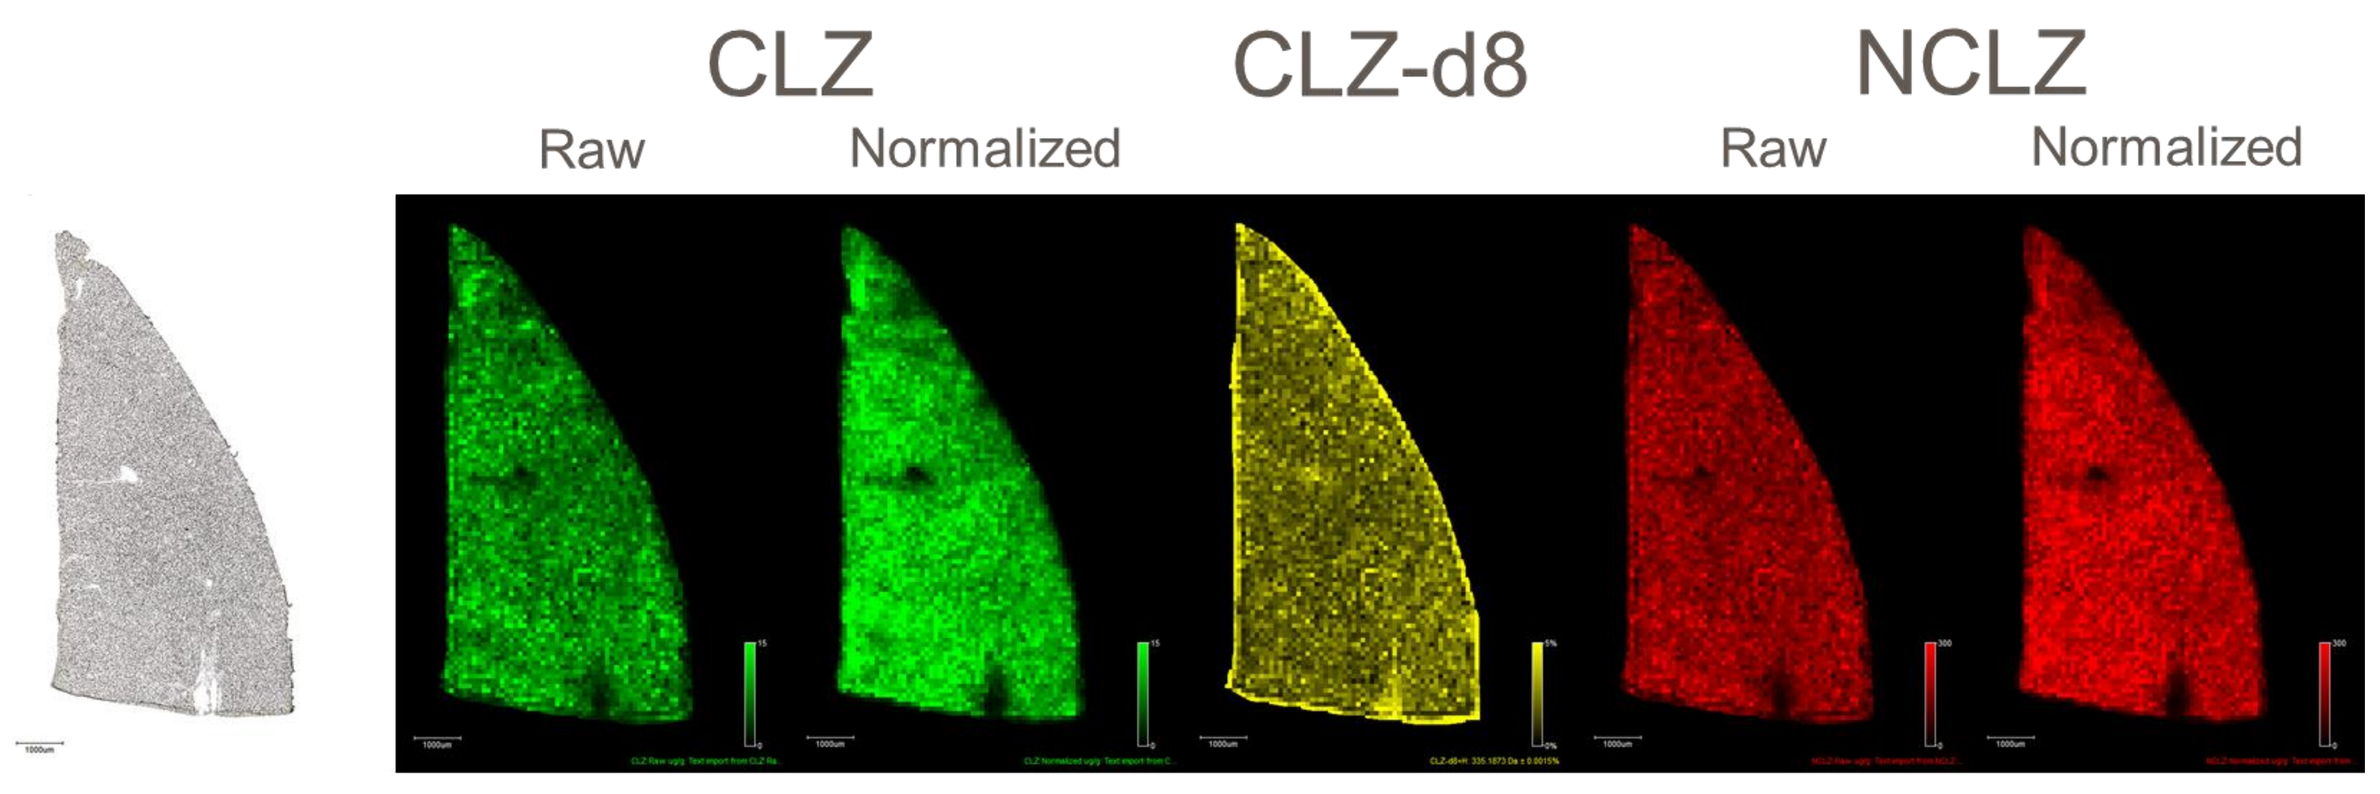
CLZ
CLZ-d8
NCLZ
Raw
Normalized
Raw
Normalized

Figure 7: Impact of Normalization on Image Quality
Normalization of both clozapine and norclozapine to the deuterated internal standard clozapine-d8 had little impact on the variability of the analyte signal within the tissue. Largely, the normalization highlighted areas with reduced suppression such as large blood vessels.

Matrix Application

Each analyst used the TM Sprayer and the same matrix application method. **Figure 8** shows a plot of matrix density against concentration of CLZ and NCLZ. The calculated matrix densities were relatively similar across the three individual systems used owing to the reproducibility of this automated matrix application approach. There was no obvious impact on the quantification within the matrix densities which were observed.

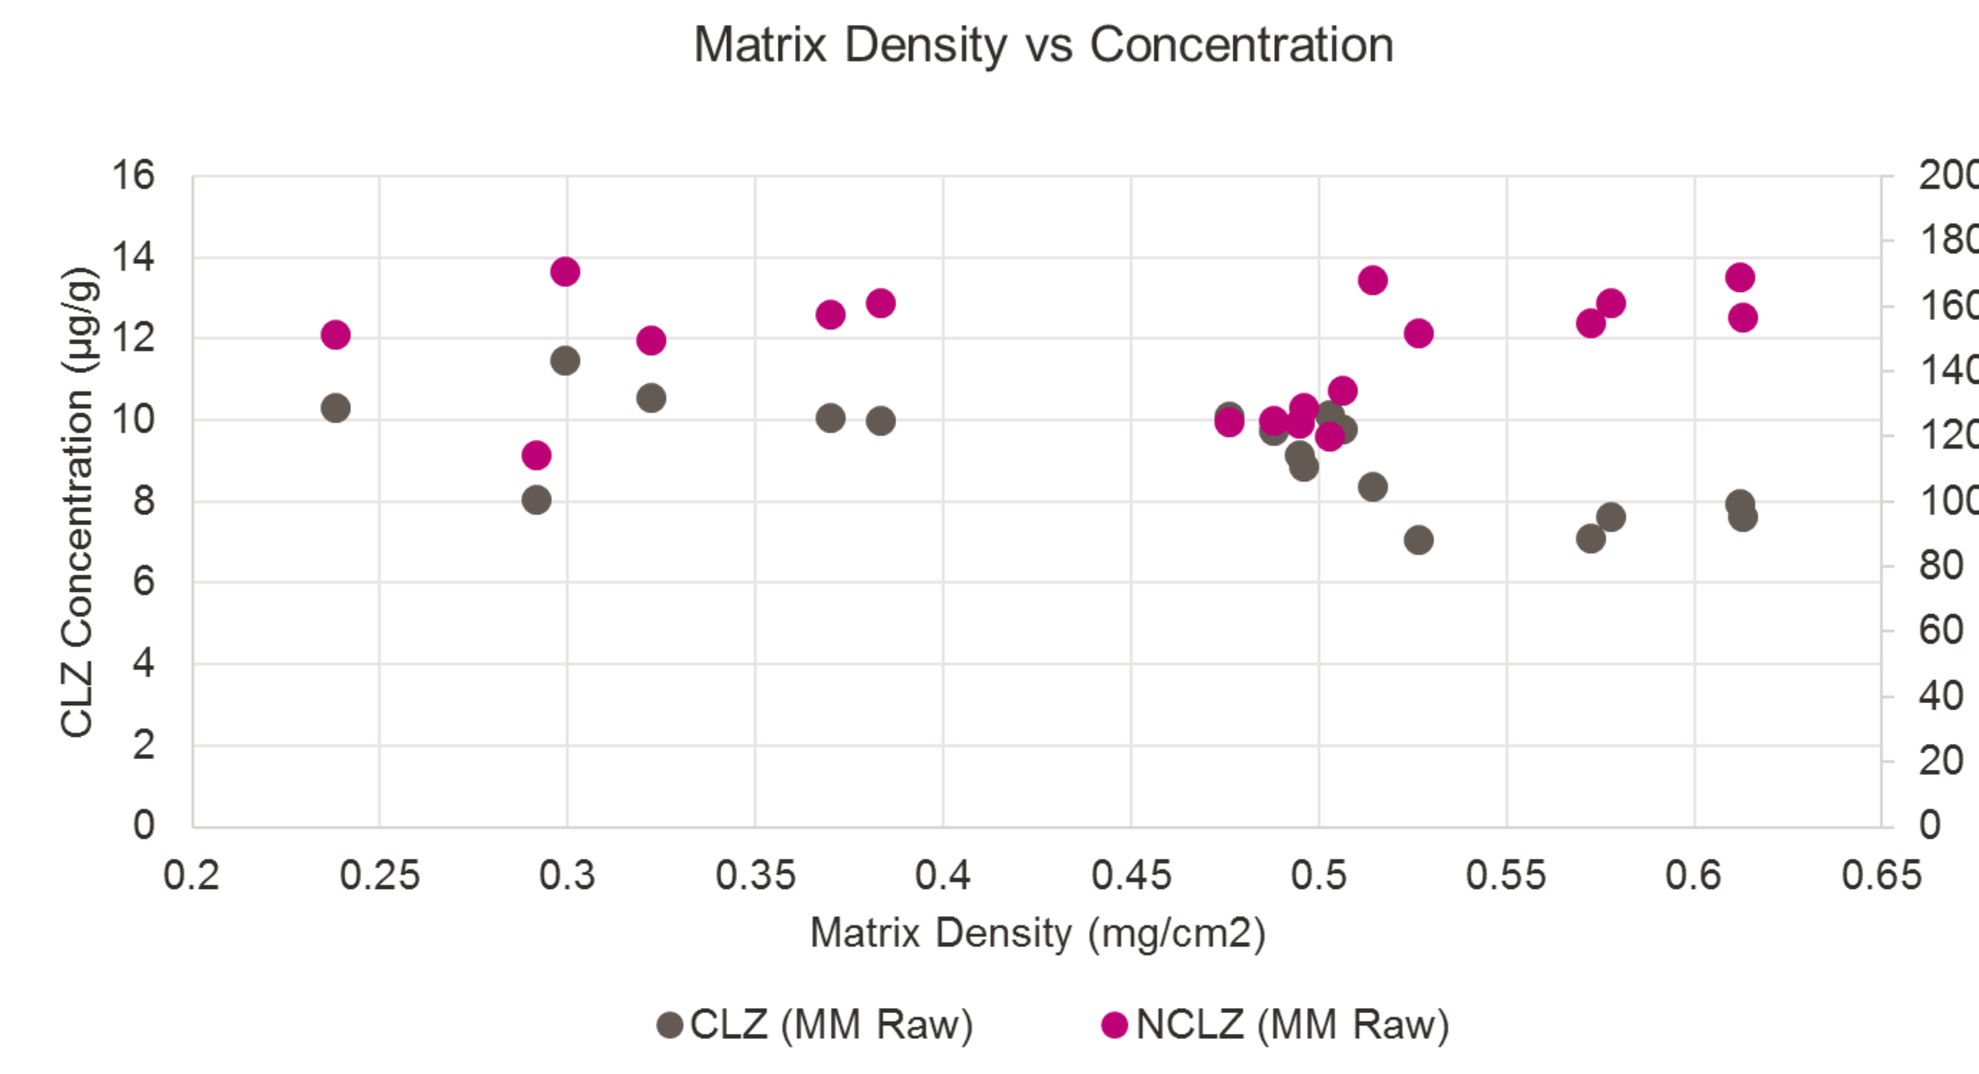
Matrix Density vs Concentration
CLZ Concentration (µg/g)
NCLZ Concentration (µg/g)
Matrix Density (mg/cm2)
CLZ (MM Raw)
NCLZ (MM Raw)

Figure 8: Matrix Density and IMS Quantification
The matrix density is plotted against the concentration for clozapine (left axis) and norclozapine (right axis)

Conclusions

All and all these data are promising and portray a positive future for quantitative IMS. A fit for purpose validation of IMS quantification using more established techniques may be warranted in certain circumstances however, the accuracy and precision of the IMS data alone appears adequate. In general normalization provides only modest improvement of the mimetic tissue model but appears to be necessary for the dilution series.

Acknowledgements

The authors would like to thank Reid Groseclose and Fang Xie for their comments on the standardized protocol. Additionally, we would like to thank Hermes Licea-Perez and Clara Andonian of the GSK Bioanalysis group for performing the LC-MS/MS analysis

References

1. Rzagalinski, I.; Volmer, D. A., Quantification of low molecular weight compounds by MALDI imaging mass spectrometry – A tutorial review. *Biochimica et Biophysica Acta (BBA) - Proteins and Proteomics* **2017,** *1865* (7), 726-739.
2. Castellino, S.; Groseclose, M. R.; Wagner, D., MALDI imaging mass spectrometry: bridging biology and chemistry in drug development. *Bioanalysis* **2011,** *3* (21), 2427-2441.
3. Bonnel, D.; Legouffe, R.; Hamm, G.; Michel, C.; Hochart, G.; Stauber, J.; Pamelard, F., Toward quantitative imaging mass spectrometry. *Curr. Trends Mass Spectrom* **2012**, 1-7.
4. Porta, T.; Lesur, A.; Varesio, E.; Hopfgartner, G., Quantification in MALDI-MS imaging: what can we learn from MALDI-selected reaction monitoring and what can we expect for imaging? *Analytical and Bioanalytical Chemistry* **2015,** *407* (8), 2177-2187.
5. Groseclose, M. R.; Castellino, S., A Mimetic Tissue Model for the Quantification of Drug Distributions by MALDI Imaging Mass Spectrometry. *Analytical Chemistry* **2013,** *85* (21), 10099-10106.
6. Barry, J. A.; Groseclose, M. R.; Fraser, D. D.; Castellino, S. Revised Preparation of a Mimetic Tissue Model for Quantitative Imaging Mass Spectrometry *Protocol Exchange* [Online], 2018. http://dx.doi.org/10.1038/protex.2018.104.

All studies were conducted in accordance with the GSK Policy on the Care, Welfare and Treatment of Laboratory Animals and were reviewed the Institutional Animal Care and Use Committee either at GSK or by the ethical review process at the institution where the work was performed.

Imaging Mass Spectrometry Society & OurCon Joint Conference November 2018